SepINRIA v1.7.2: Multiple Sclerosis Brain Visualisation, Comparison and Analysis Software

http://www-sop.inria.fr/asclepios/software/SepINRIA/

Erik Pernod[1], Jean-Christophe Souplet[1], Mikael Cohen[2], Nicolas Toussaint[1], Christine Lebrun[2], Grégoire Malandain[1]

1. Asclepios Research Team – INRIA Sophia-Antipolis, France 2. Neurology Department, CHU Pasteur, Nice, France

World Congress on Treatment and Research in Multiple Sclerosis
Montréal 2008

1. Objectives

To provide clinicians with tools allowing to analyze Multiple Sclerosis (MS) brain MRI:

- Works on a convenient database.
- Visualisation of images in 2D and/or 3D.
- Quantitative lesion burden evaluation (manually or automatically).
- Brain atrophy evaluation (manually or automatically).
- Comparison of different sequences or of binary segmentations.

2. Data Management & Visualisation

The database interface allows to:

- Import images from DICOM and other formats.
- Rotate/Flip images.
- Export results in DICOM or other formats (e.g. the segmentations).

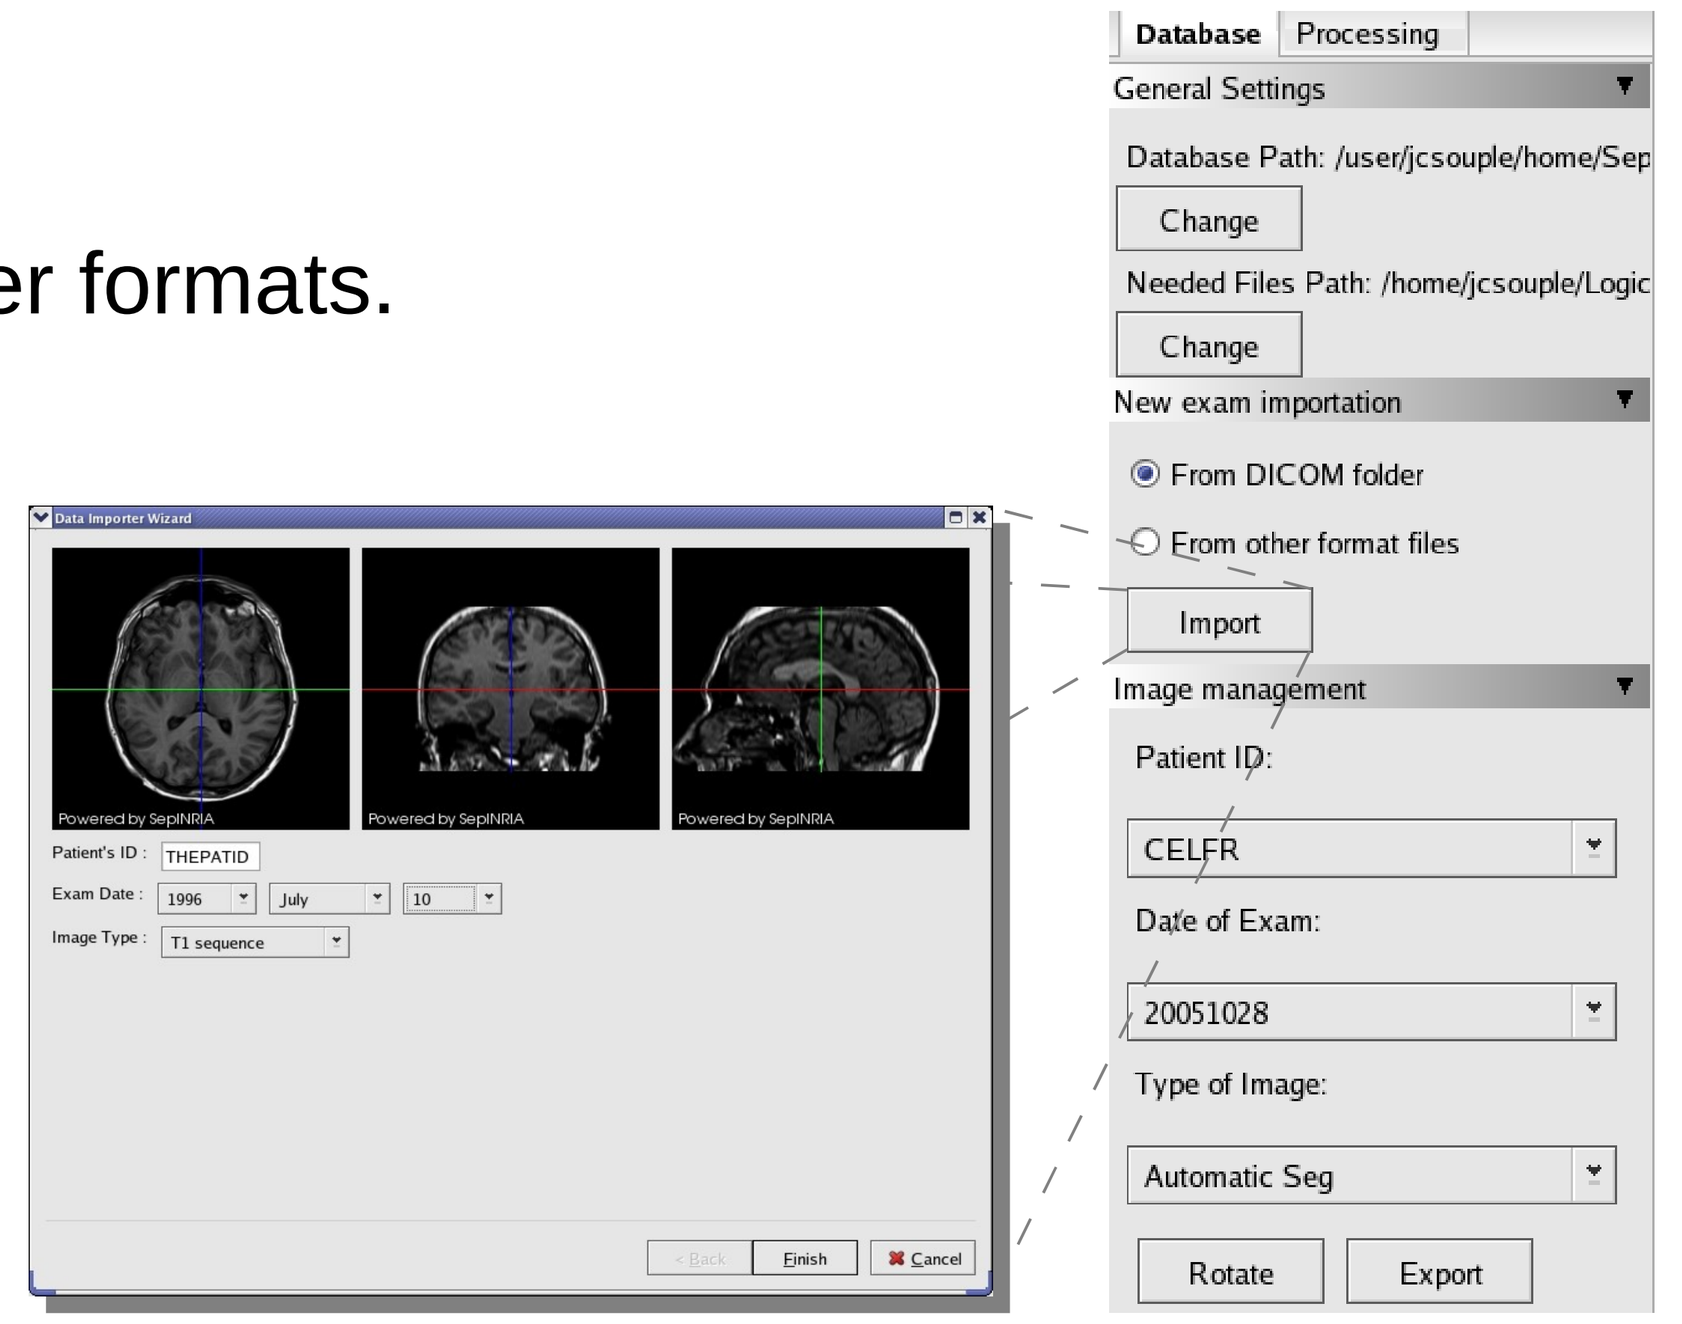

Database
Processing
General Settings
Database Path: /user/jcsouple/home/Sep
Change
Needed Files Path: /home/jcsouple/Logic
Change
New exam importation
From DICOM folder
From other format files
Import
Image management
Patient ID:
CELFR
Date of Exam:
20051028
Type of Image:
Automatic Seg
Rotate
Export

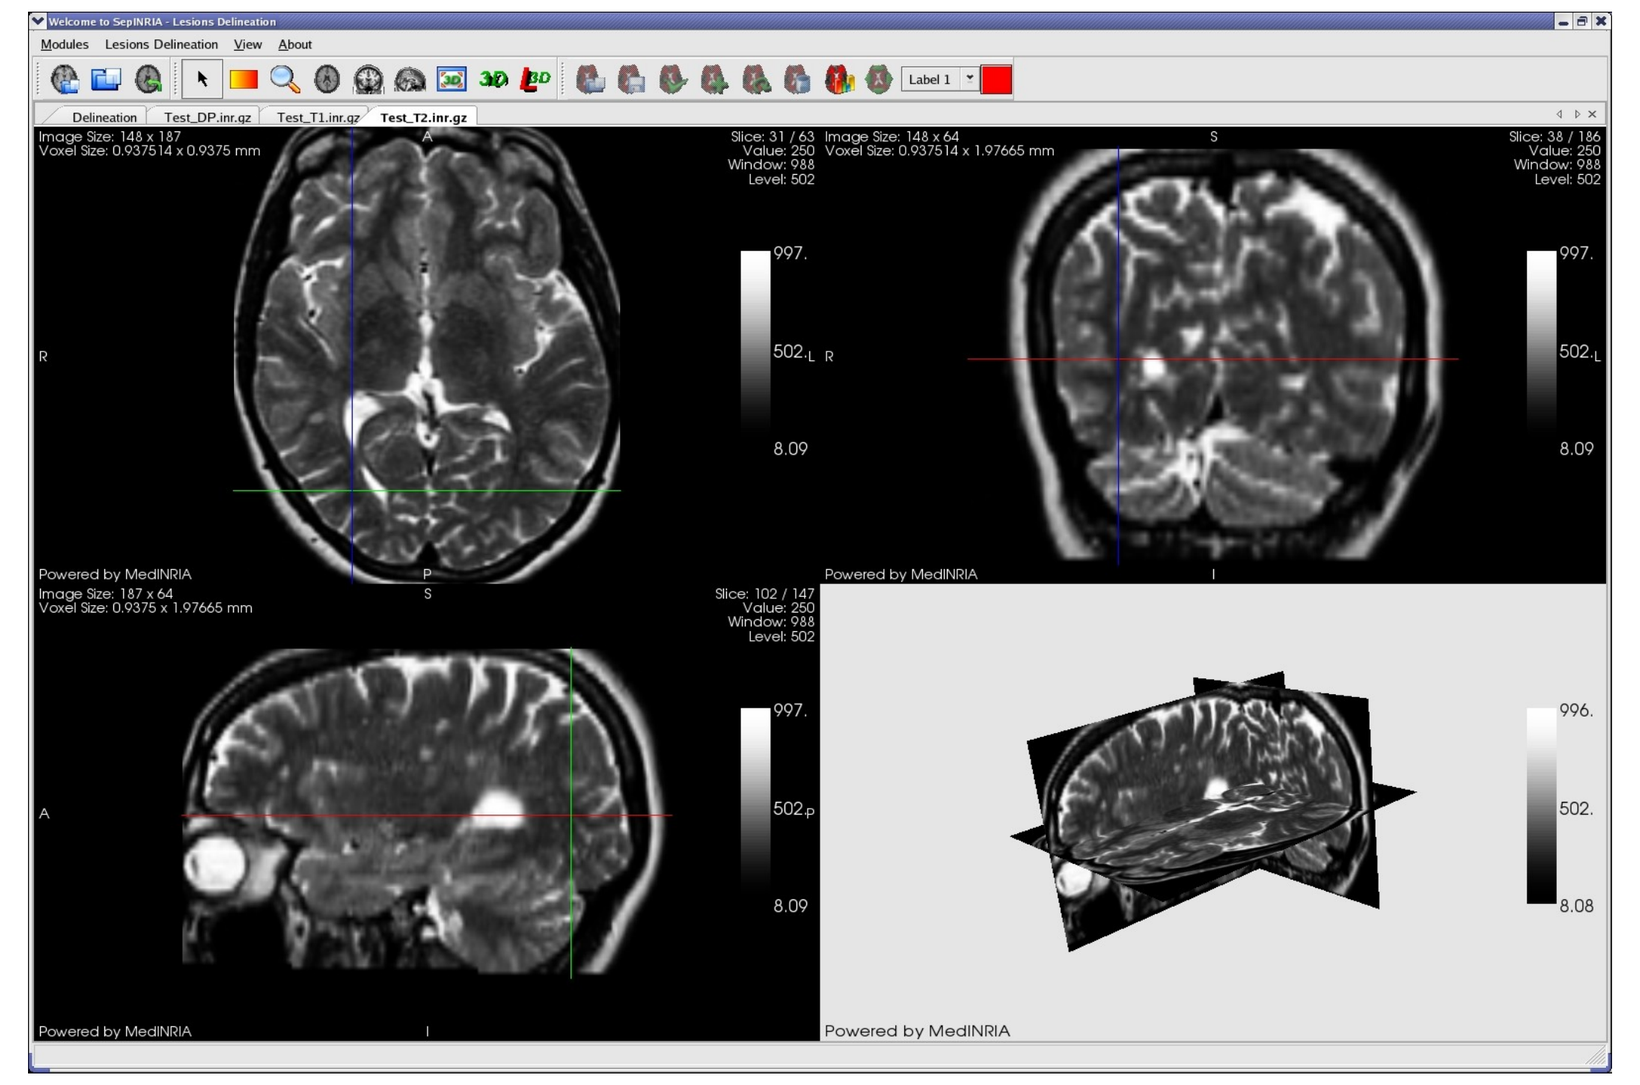

Different interactions are possible:

- Display in 2D/3D.
- Zoom.
- Windowing.
- Volume rendering.

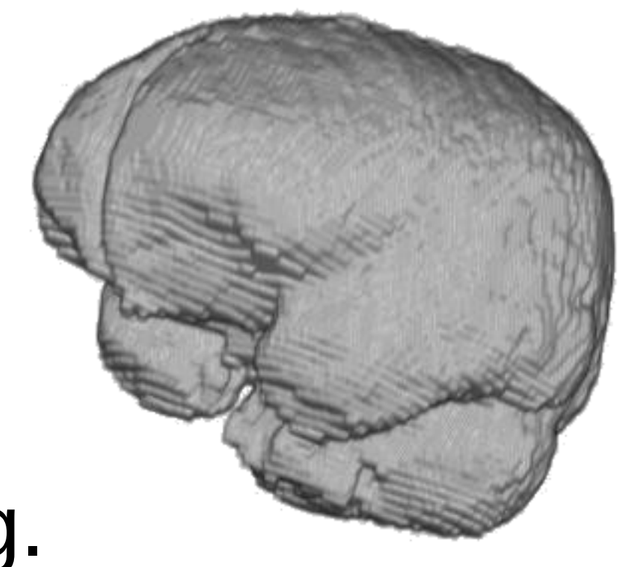

3. Lesion Segmentation

Manually:

- Drawing slice by slice.
- The drawing of a slice can be used for the following slice.

Semi-automatically:

- Click in the center of a lesion.
- The segmentation is achieved thanks to a 3D region growing.

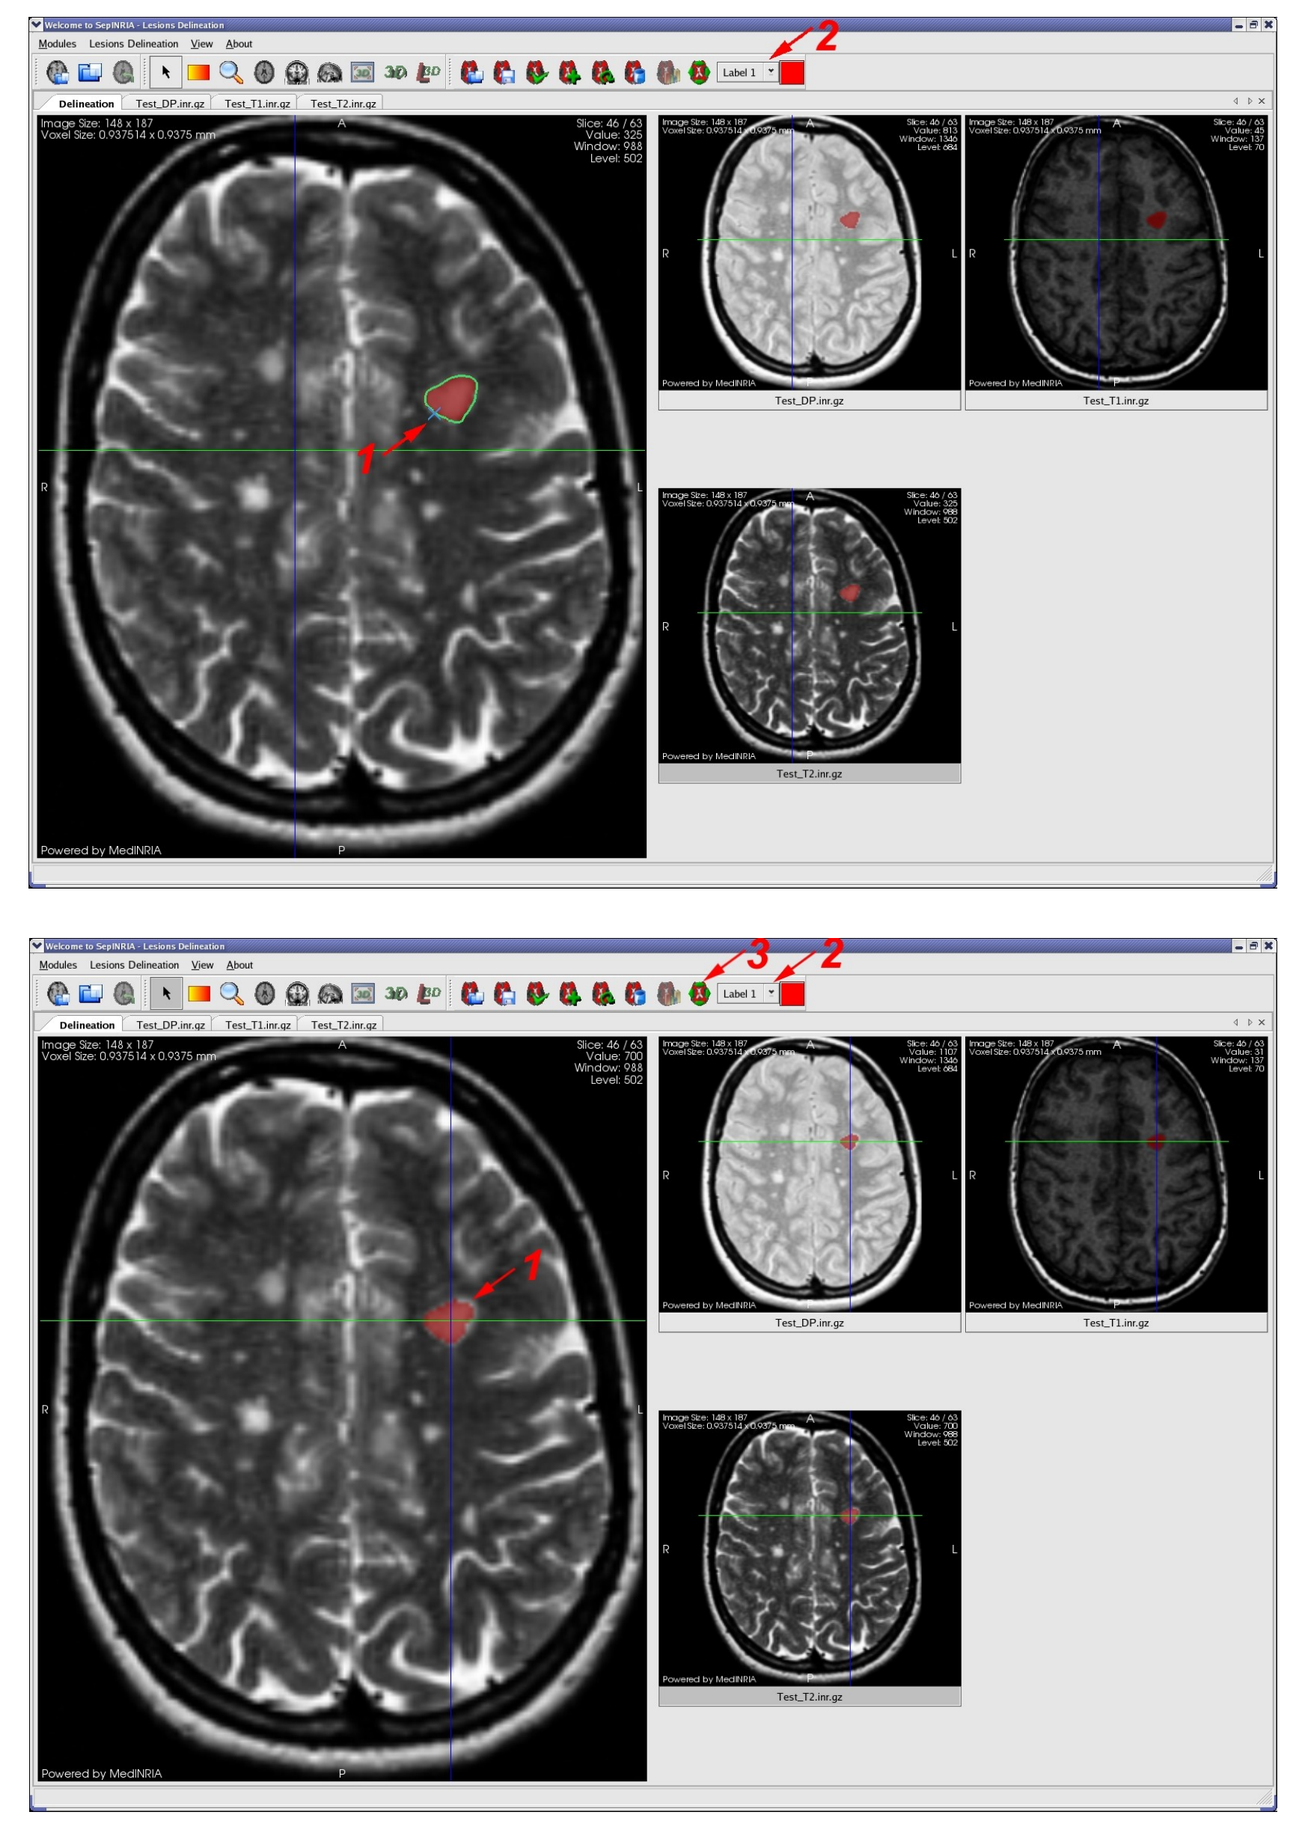

Automatically:

- From four sequences (T2, PD, T1, T2-FLAIR).
- Images normalization.
- Segmentation of WM, GM, CSF taking into consideration partial volume effects.

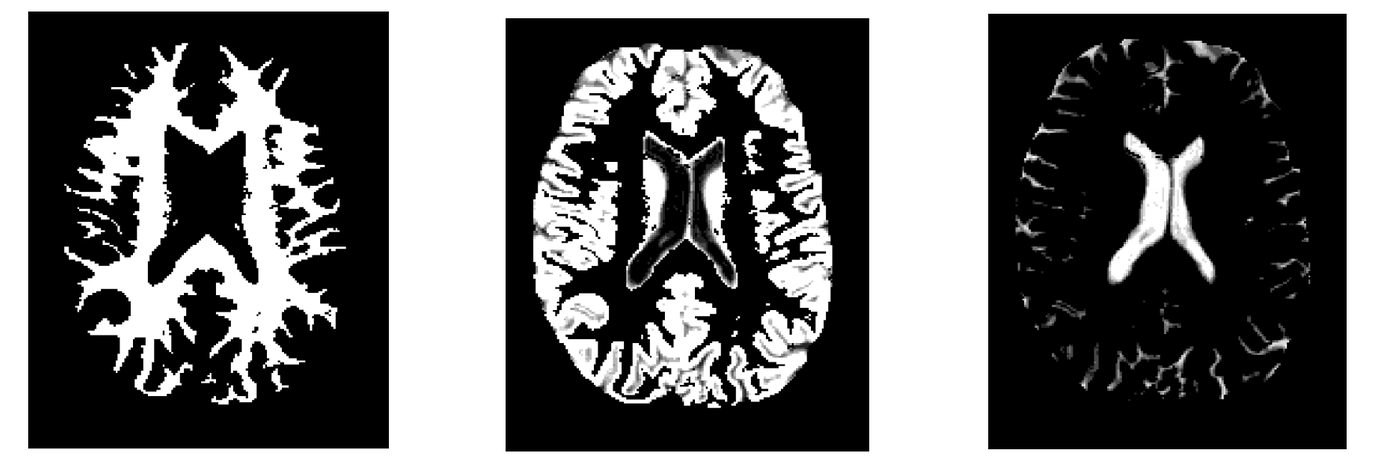

- Segmentation of the lesions.
- Lesion burden computation.

Results:

- Visualisation in 2D or 3D of the lesions.
- Lesion burden computed from segmentations.
- Segmentations saved and exported.
- A quantitative comparison of two segmentations.

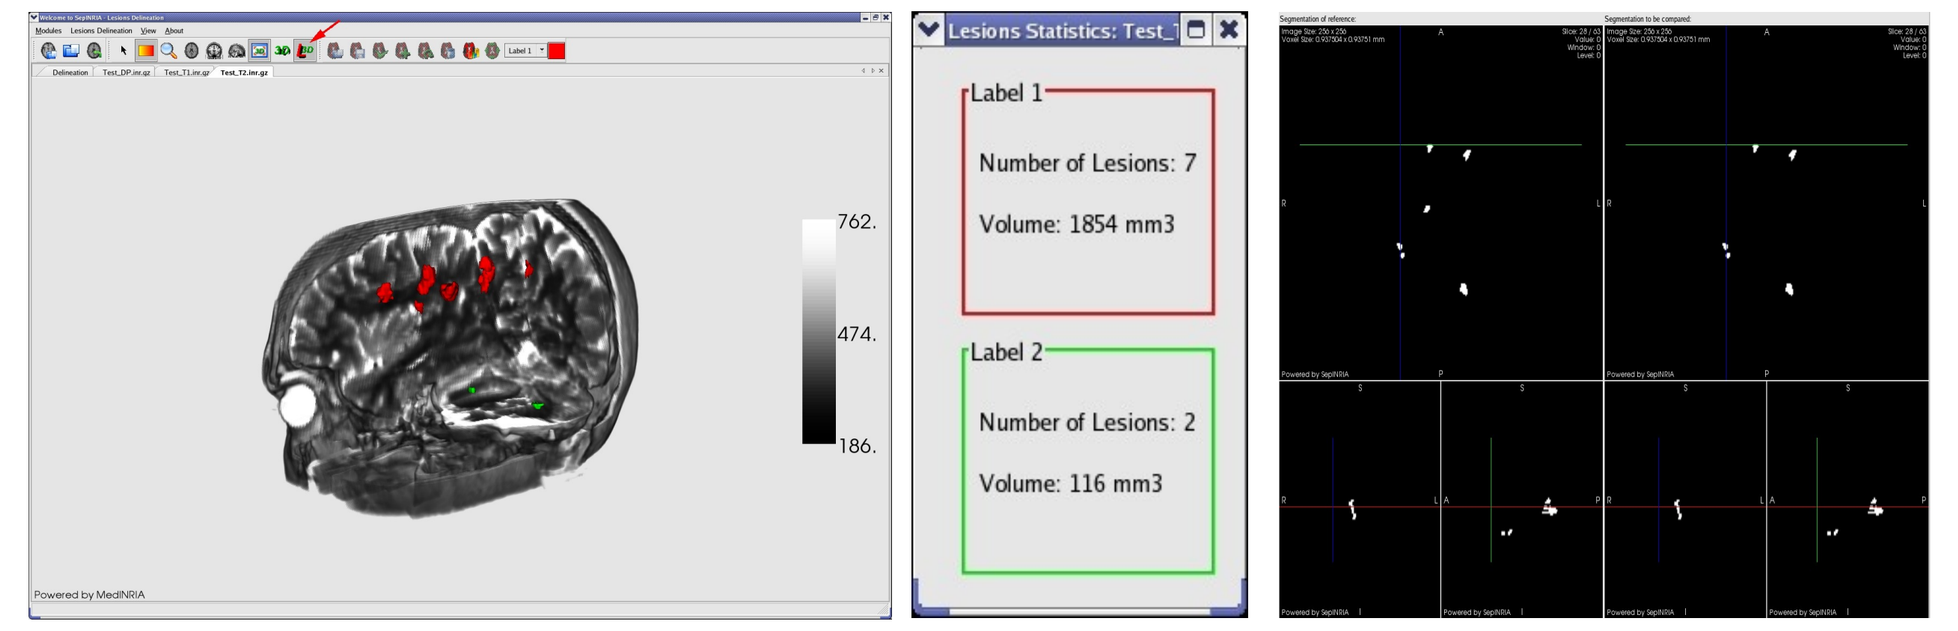

Label 1
Number of Lesions: 7
Volume: 1854 mm3
Label 2
Number of Lesions: 2
Volume: 116 mm3

4. Atrophy Evaluation

2D linear manual measurements:

- Automatic images aligning on an image of reference. Reference slices are saved into memory.
- Selection of the measurement:
 - Brain width,
 - Lateral ventricles width,
 - Third ventricle width.
- Automatic display of the corresponding 2D slice.
- Click to move landmarks.
- Save the measurement.

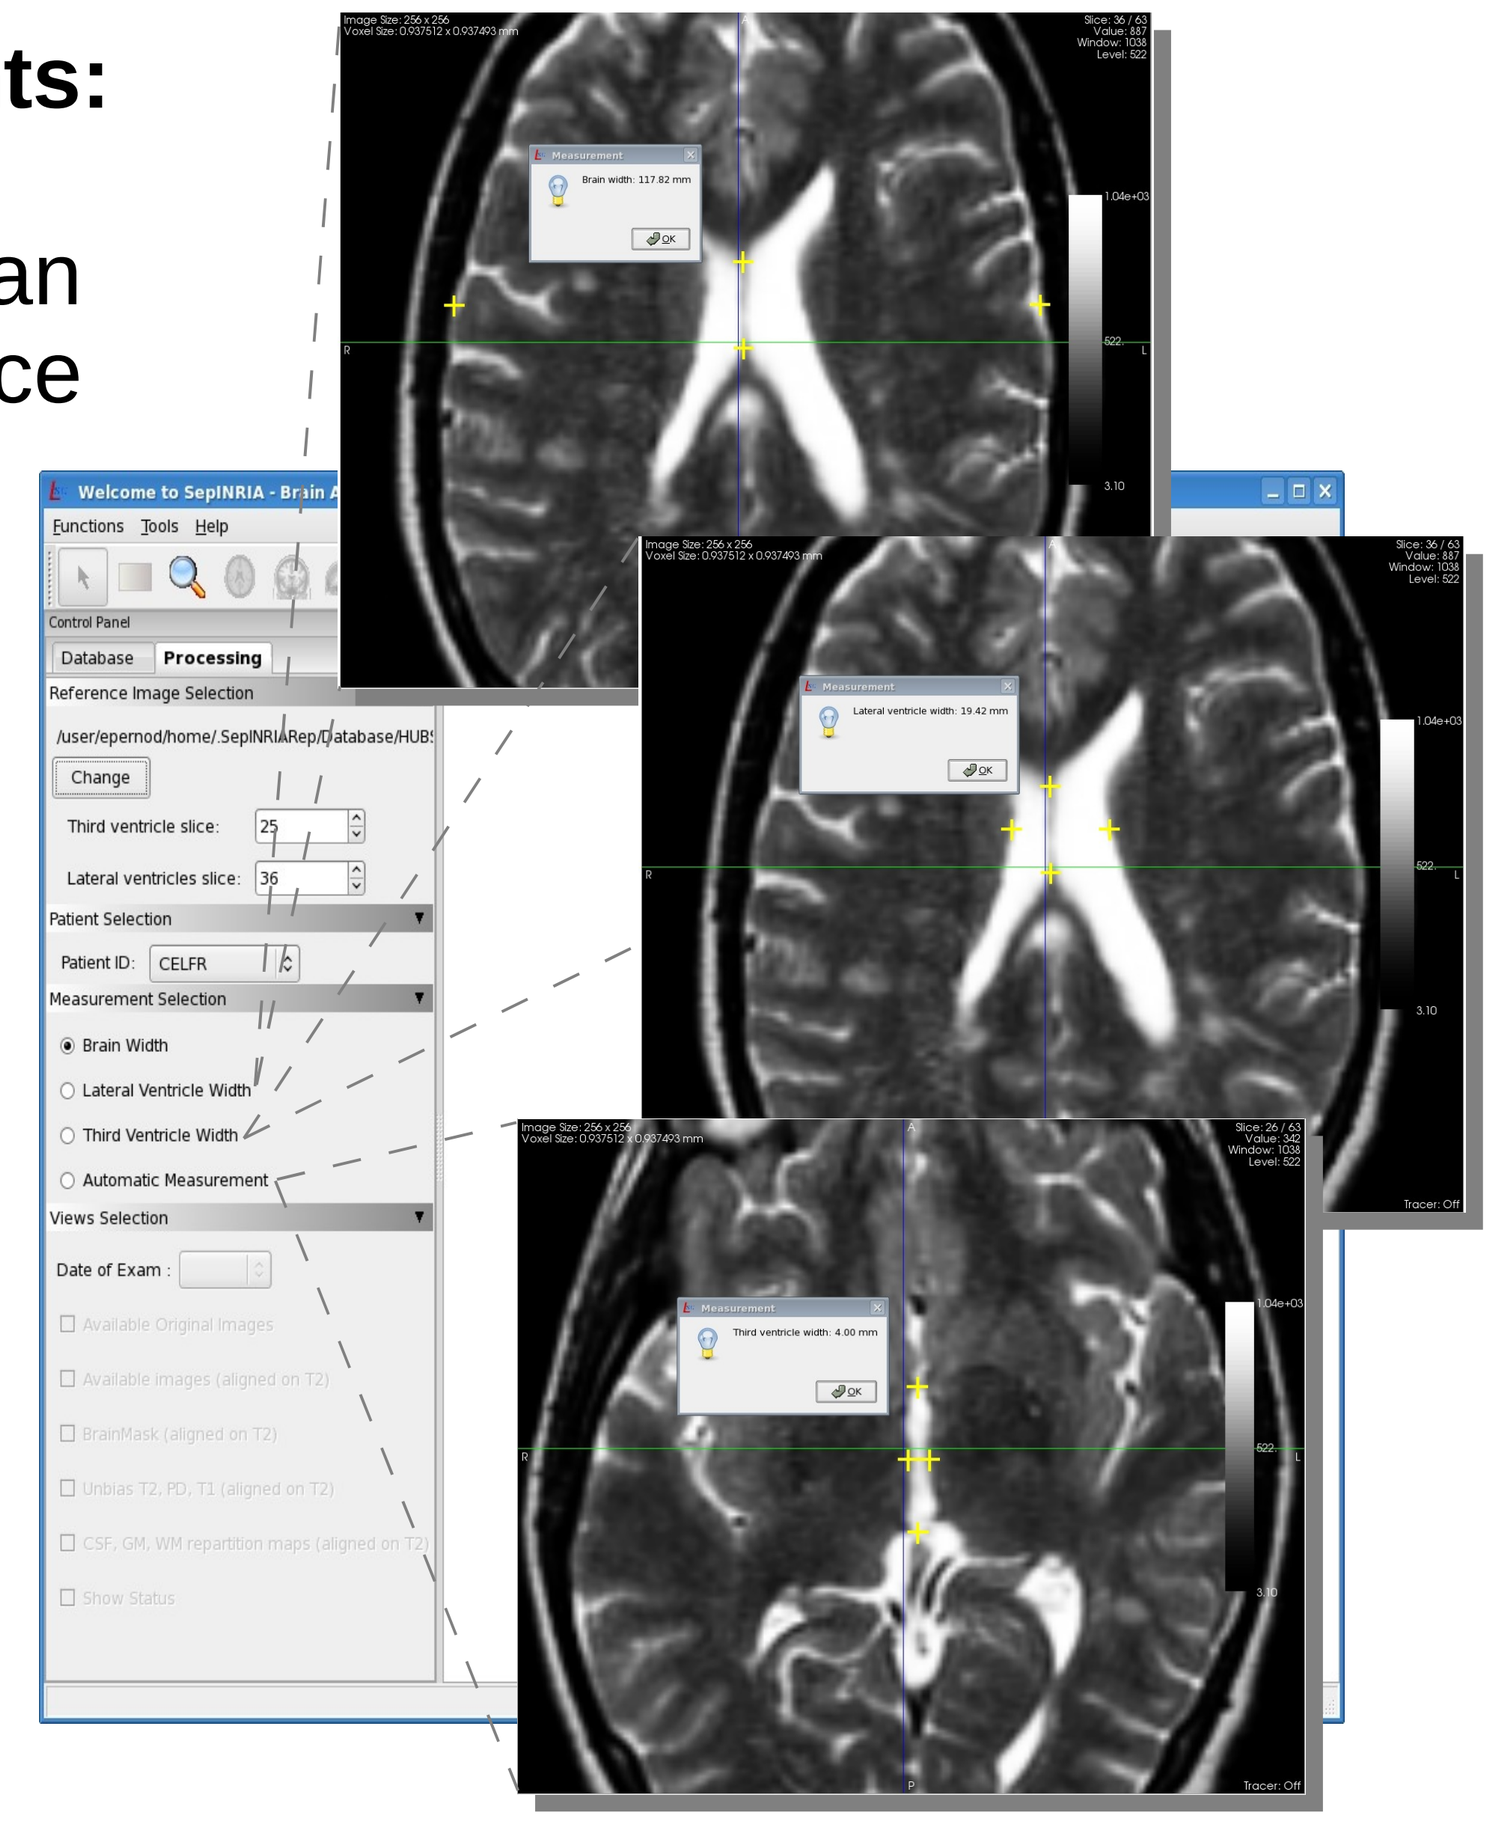

Automatically:

- From three sequences (T2, PD, T1).
- Sequences alignment.
- Segmentation of the brain.
- Intensity inhomogeneities correction.
- Segmentation of WM, GM, CSF taking into consideration partial volume effects.
- WM, GM, CSF volume and Brain Parenchymal Fraction (BPF) computation.

$$BPF = \frac{Volume(GM) + Volume(WM)}{Volume(GM) + Volume(WM) + Volume(CSF)}$$

Results:

- 2D linear measurements saved and exported.
- Display evolution of the BPF. This evolution reflects atrophy

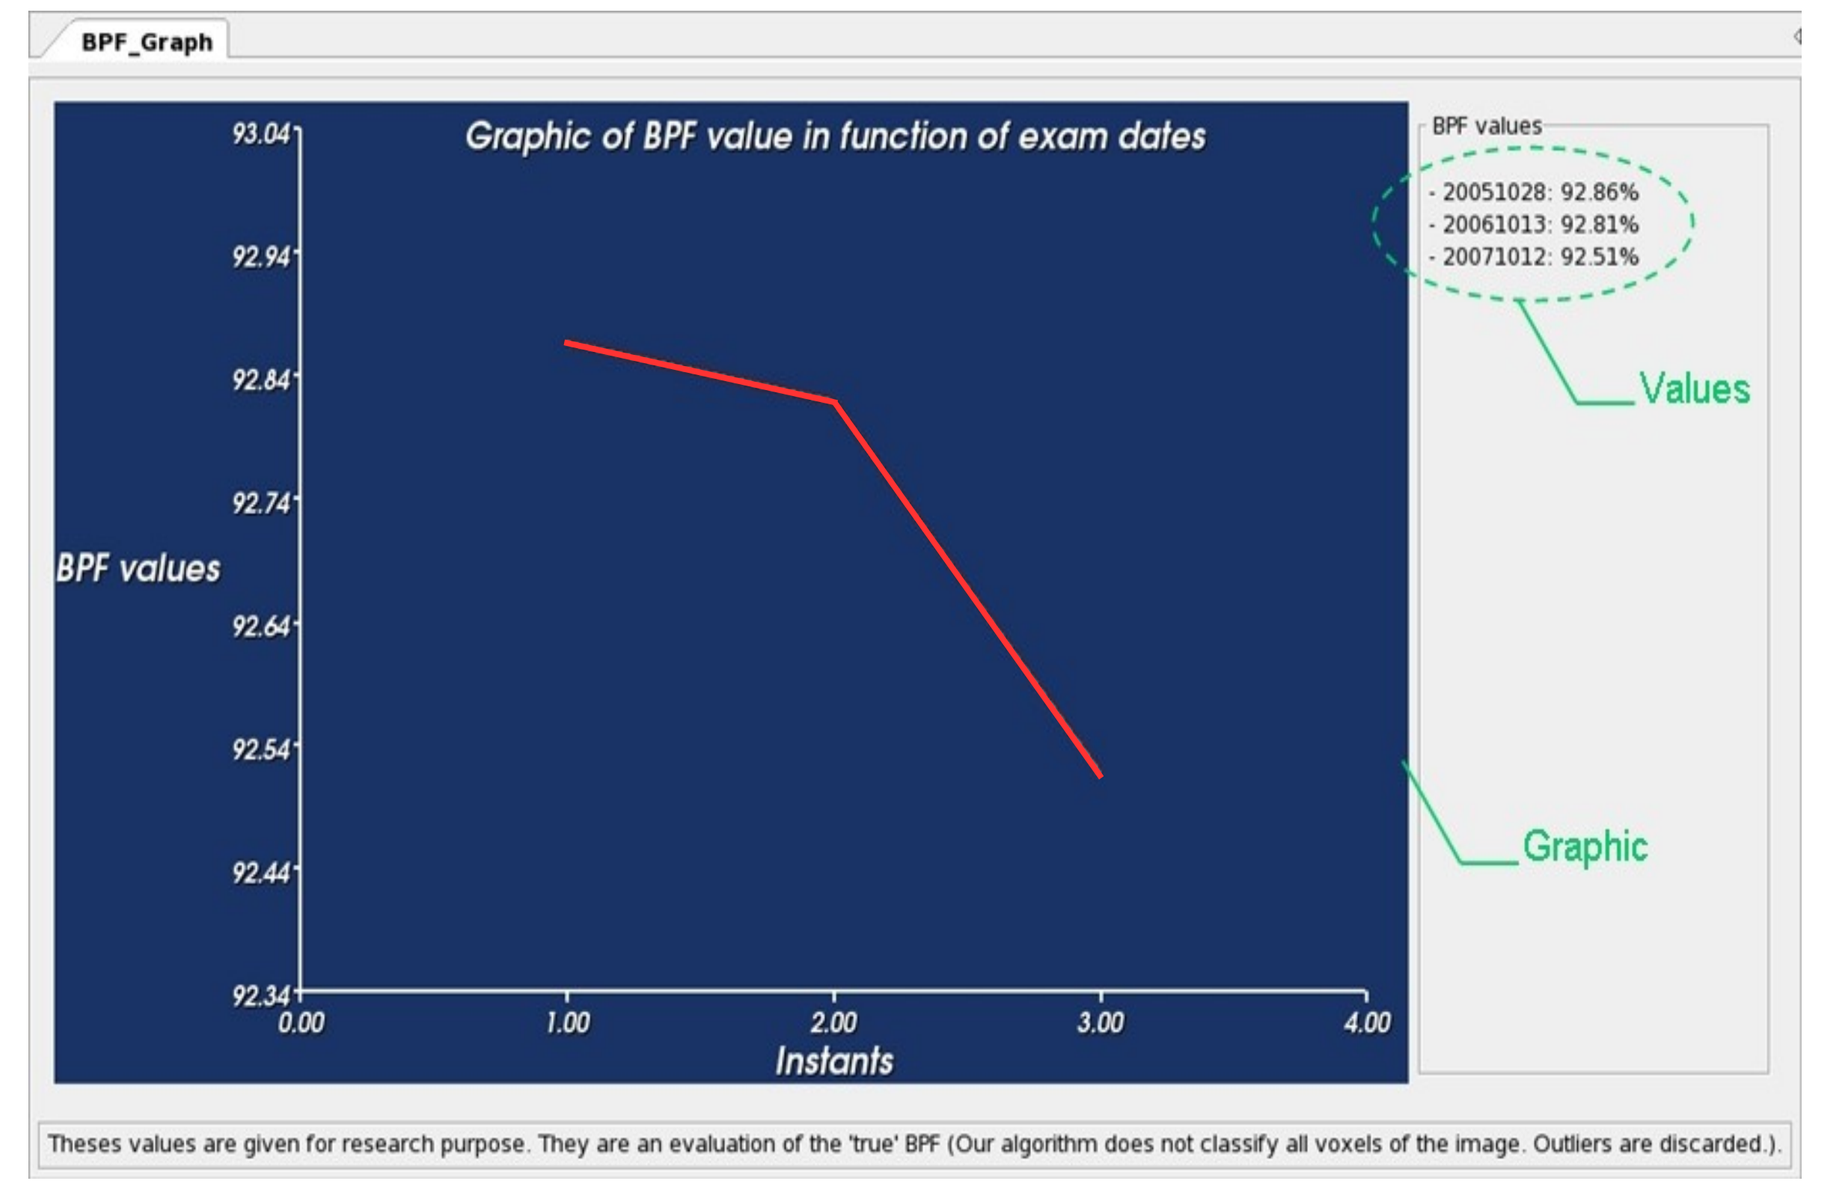

BPF_Graph
Graphic of BPF value in function of exam dates
BPF values
Instants
BPF values
- 20051028: 92.86%
- 20061013: 92.81%
- 20071012: 92.51%
Values
Graphic
Theses values are given for research purpose. They are an evaluation of the 'true' BPF (Our algorithm does not classify all voxels of the image. Outliers are discarded.).

5. Image comparison

- Automatic co-registration of images:
 - Monomodal,
 - Multimodal.
- Quantitative comparisons of two binary segmentations to assess evolutions:
 - Computation of the image difference.
 - Visualisation in the same window, side to side or by image fusion.

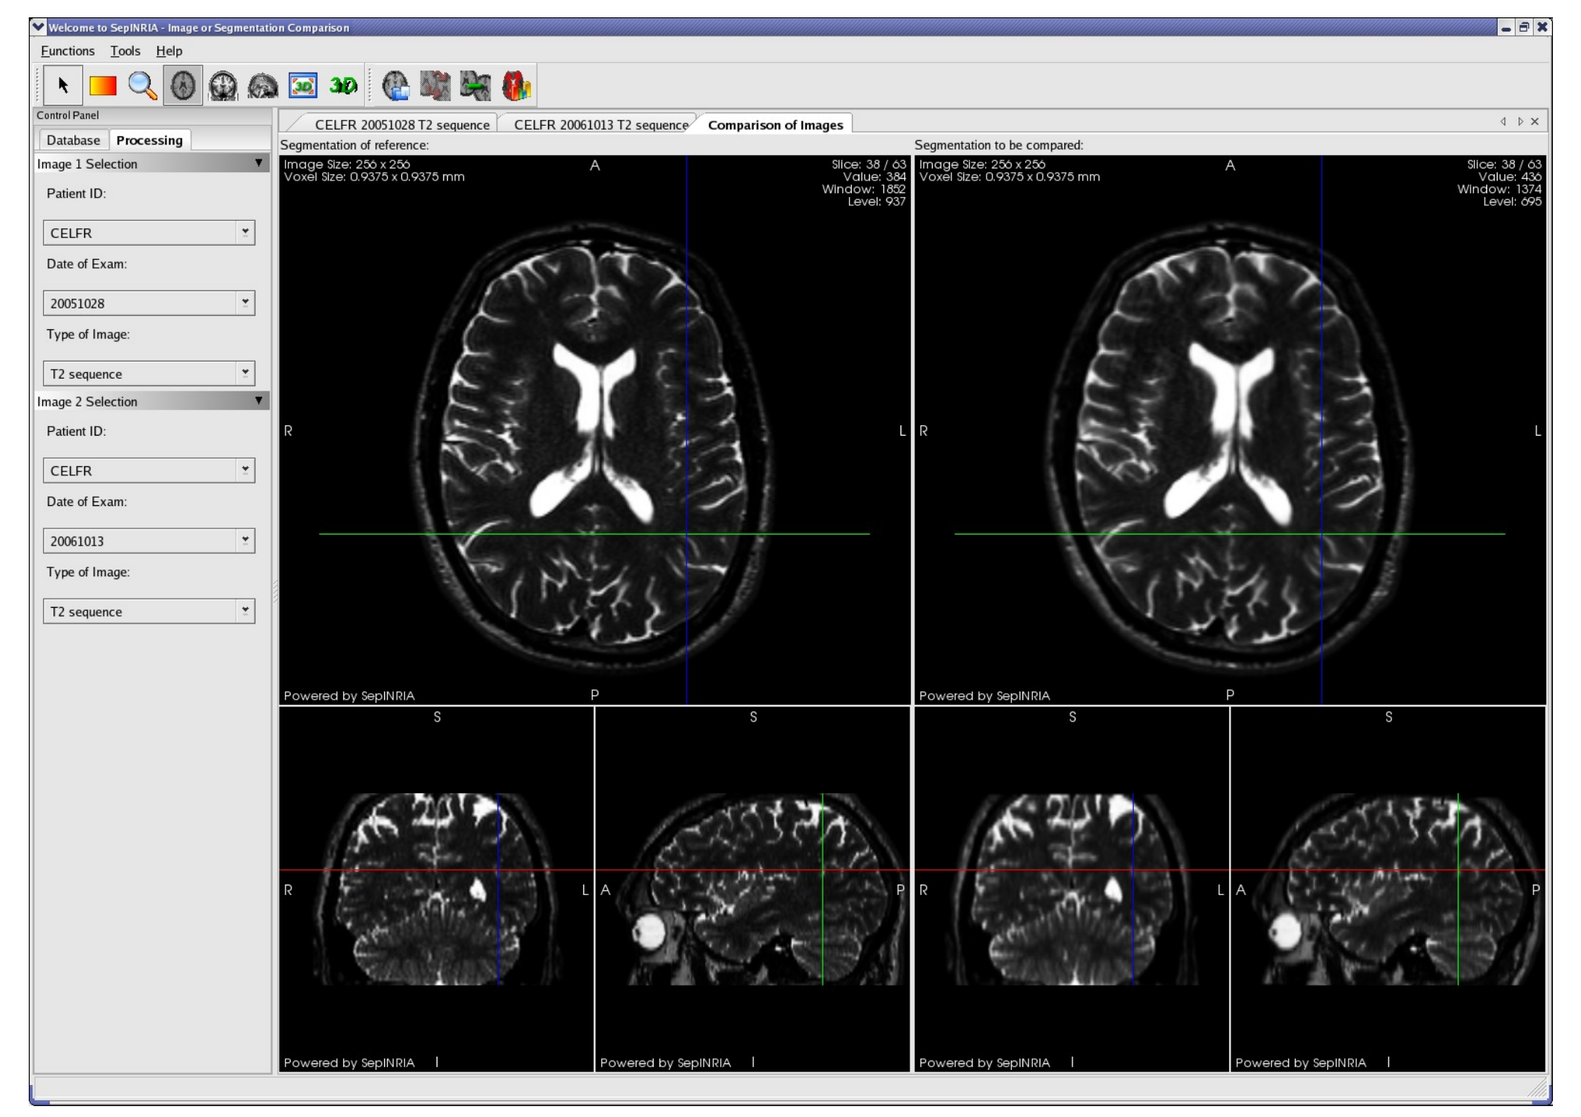

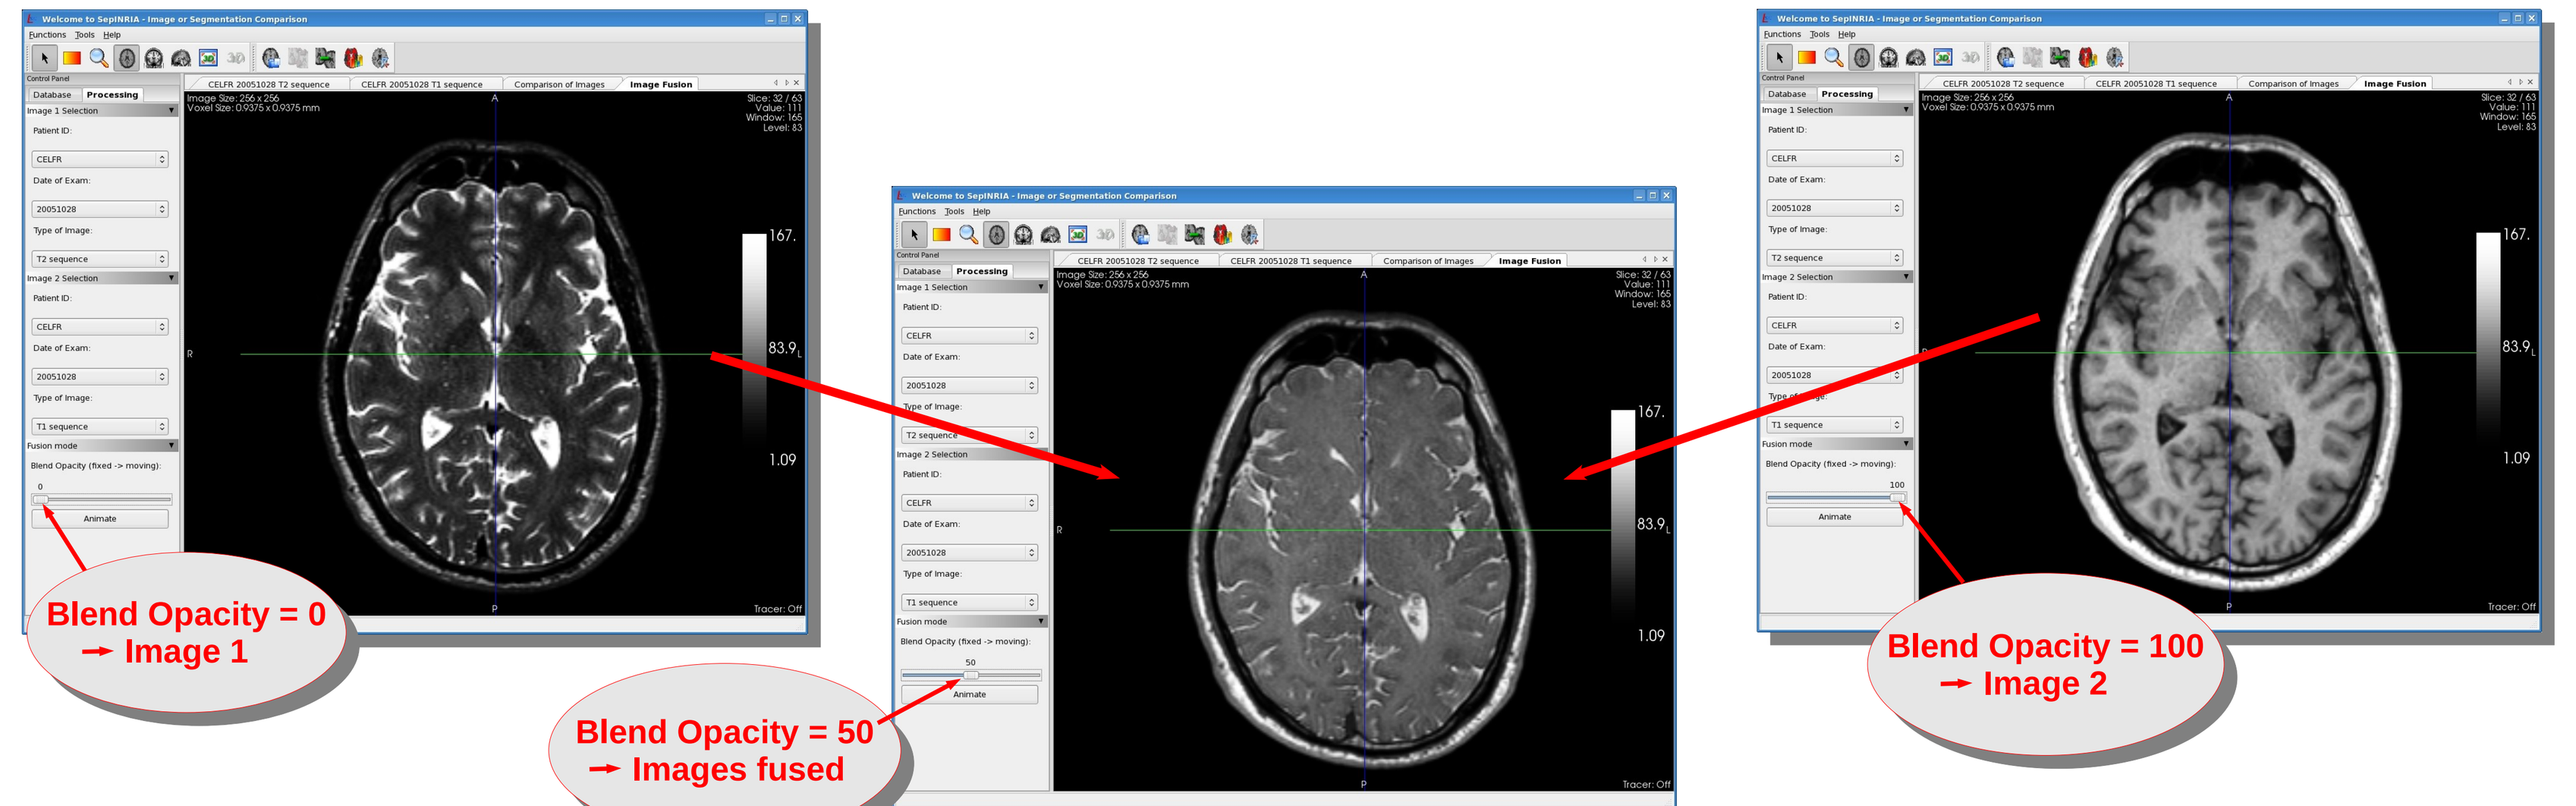

Blend Opacity = 0 → Image 1
Blend Opacity = 50 → Images fused
Blend Opacity = 100 → Image 2

6. Conclusion

- SepINRIA can be freely downloaded at:

 http://www-sop.inria.fr/asclepios/software/SepINRIA/

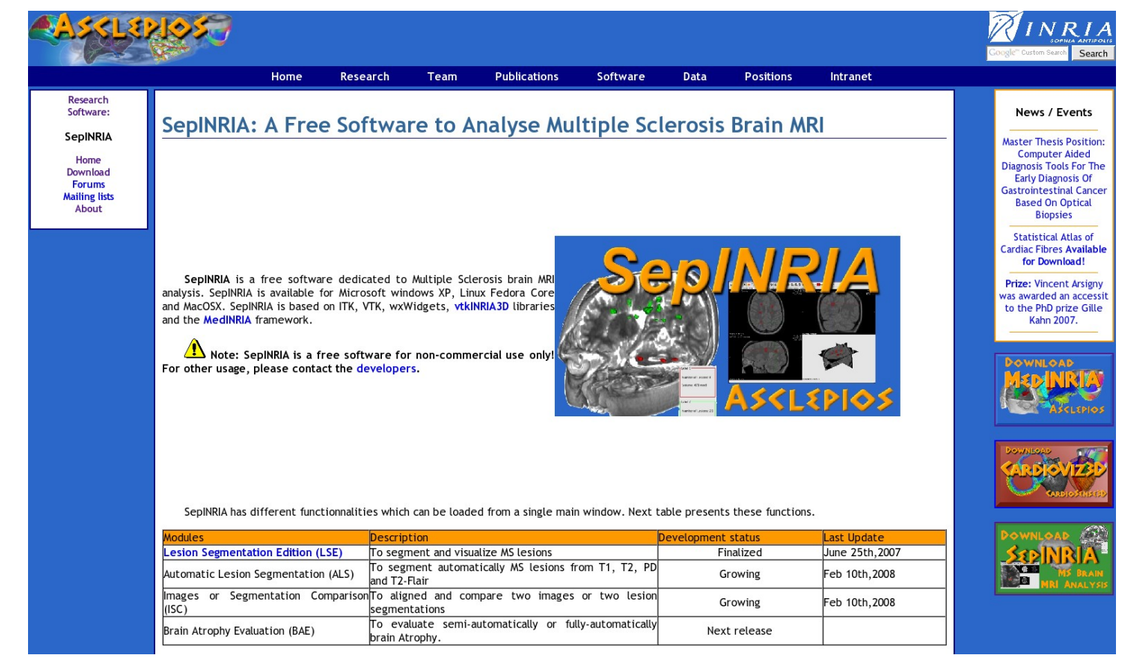

- SepINRIA is available on different platforms:
- SepINRIA is based on ITK, VTK, wxWidgets, vtkINRIA3D libraries and the MedINRIA framework.